Attention Influences the Neural Representation of Natural Scene Category

Eamon Caddigan[1,*], Audrey G. Lustig[1,], Li Fei-Fei[2], Diane M. Beck[1]

[1]University of Illinois at Urbana-Champaign, [2]Stanford University, *ecaddiga@illinois.edu

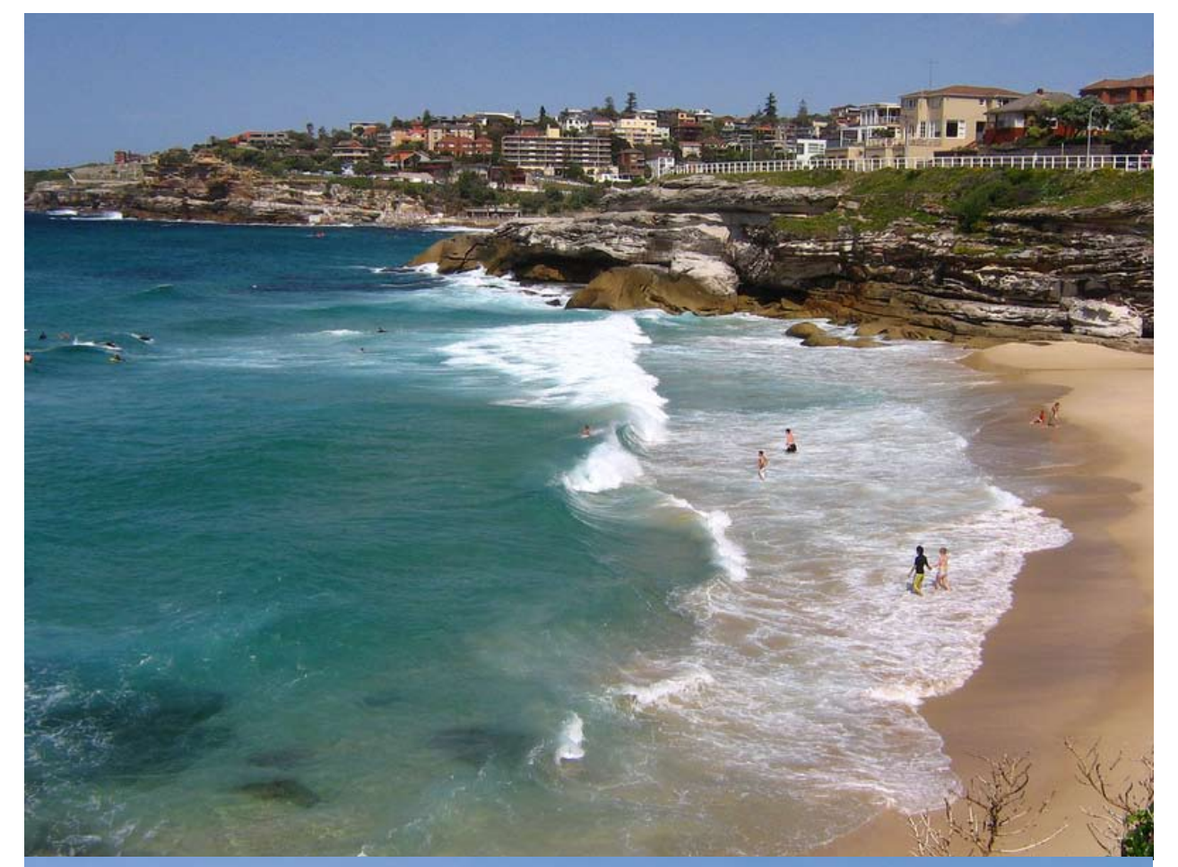
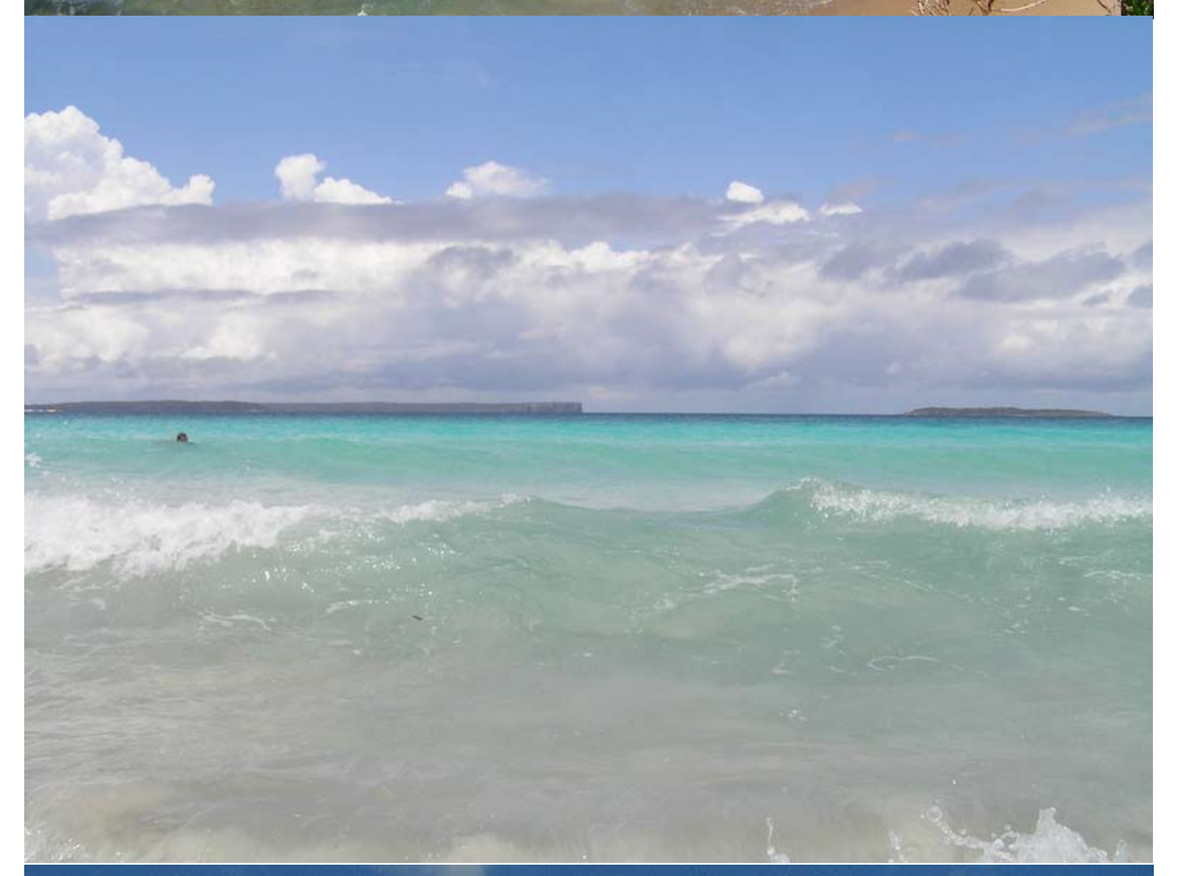
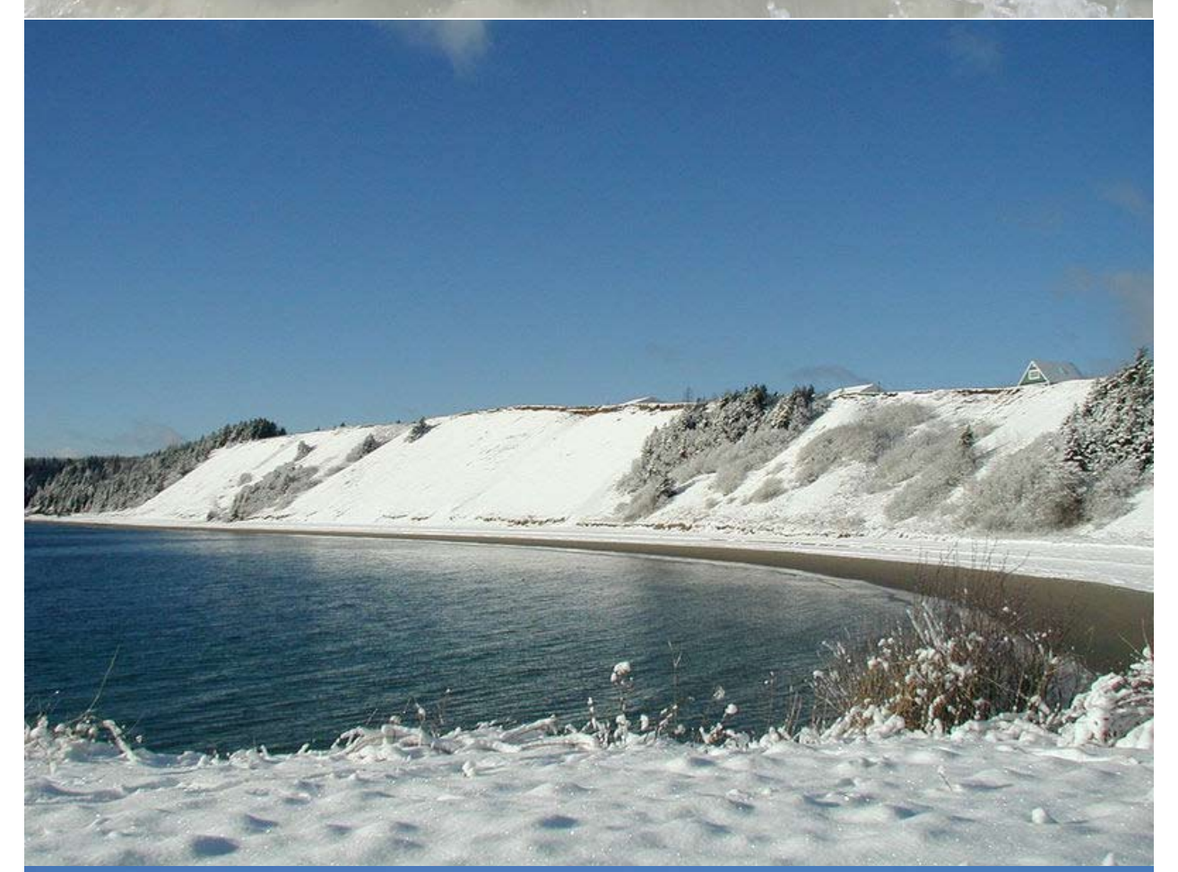
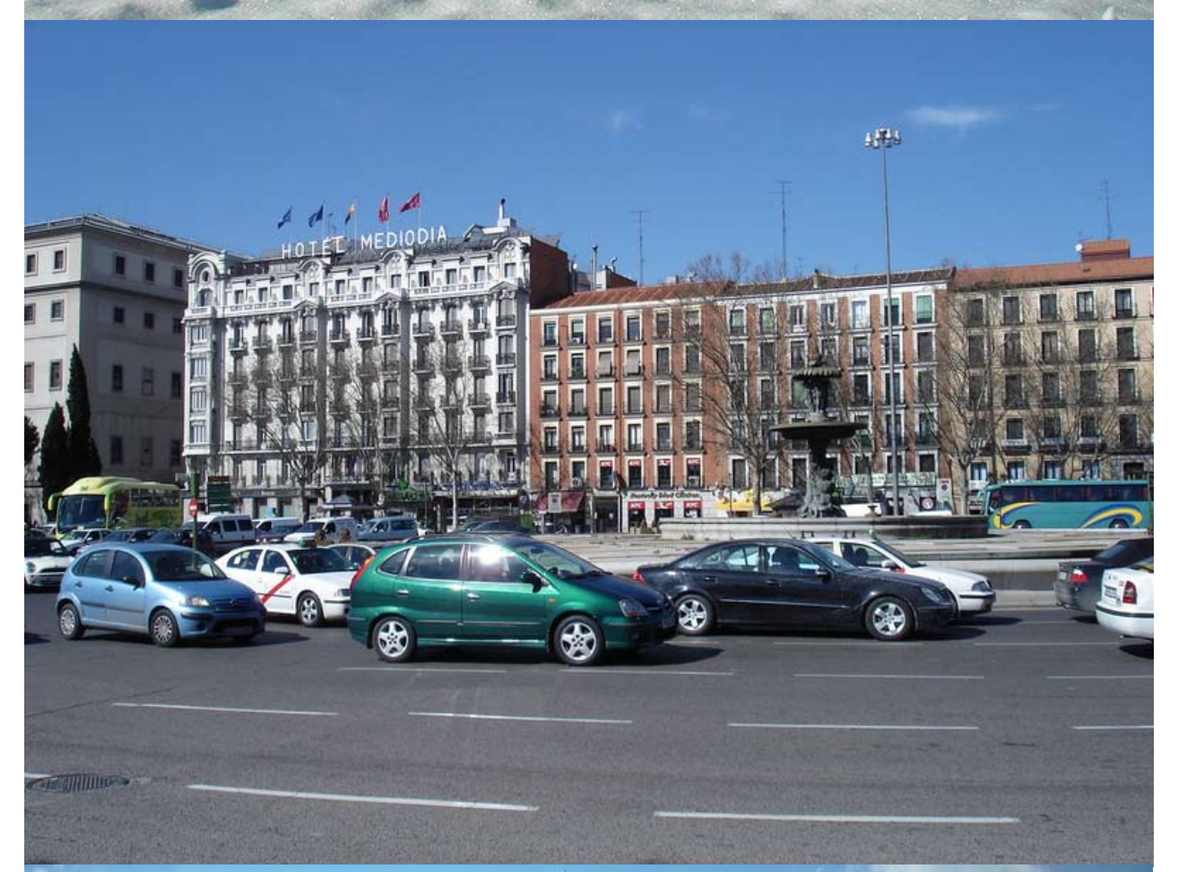
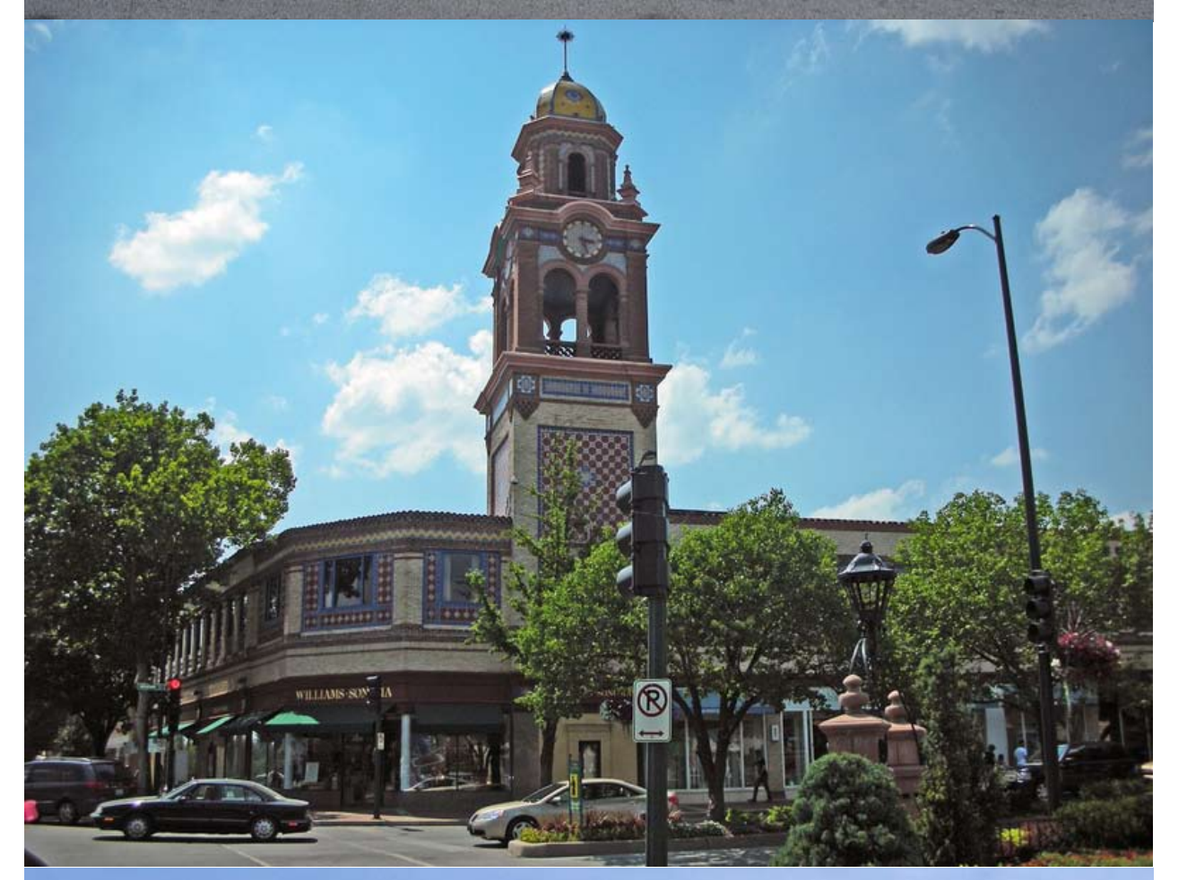
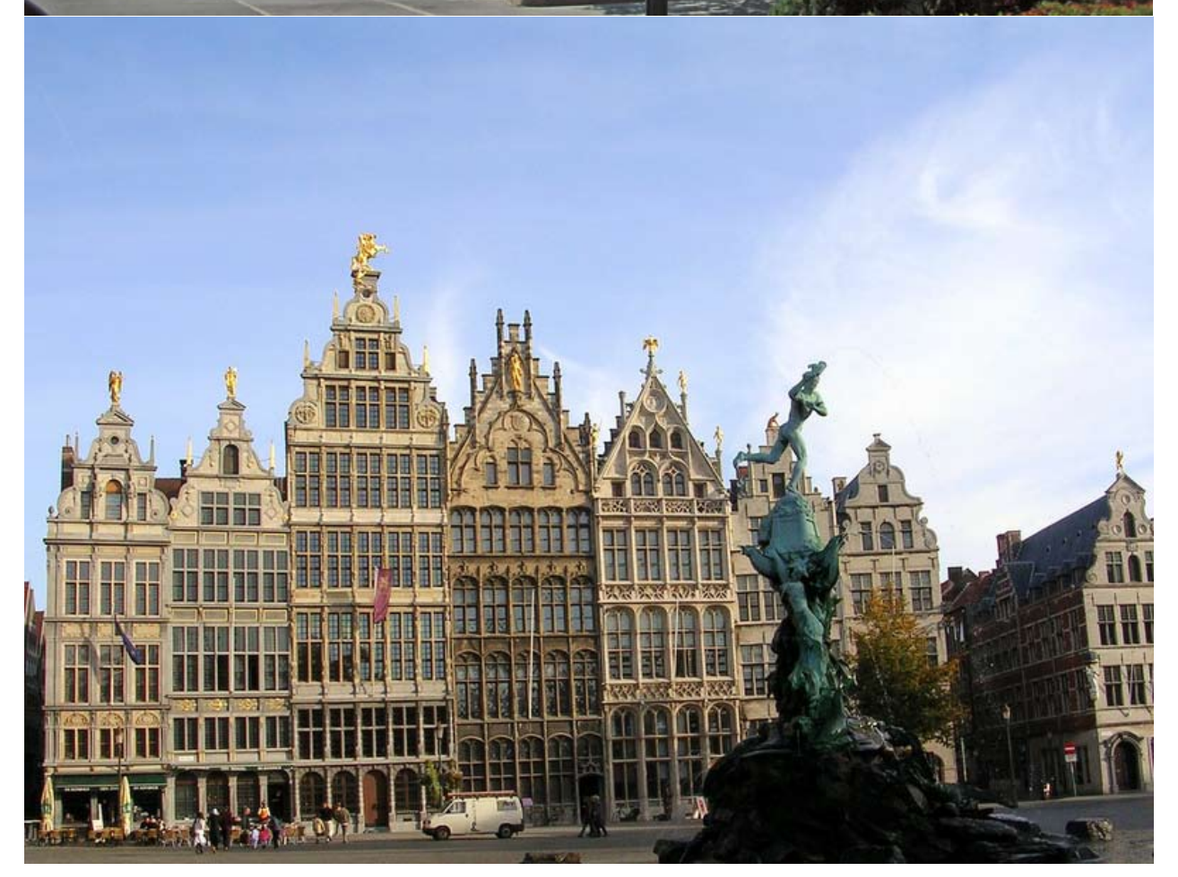

Introduction

Observers can categorize images of natural scenes while engaged in a demanding task at fixation (Li et al., 2002).

Natural scene category can be “decoded” from patterns of fMRI activity (Walther et al., 2009).

fMRI activity can identify objects in unattended scenes (Peelen, Fei-Fei, Kastner, 2009).

Can the category of unattended scenes be decoded from patterns of fMRI activity?

Does attention affect natural scene category decoding accuracy?

Parahippocampal Place Area (PPA)

Natural scene category was decoded from voxels in the PPA.

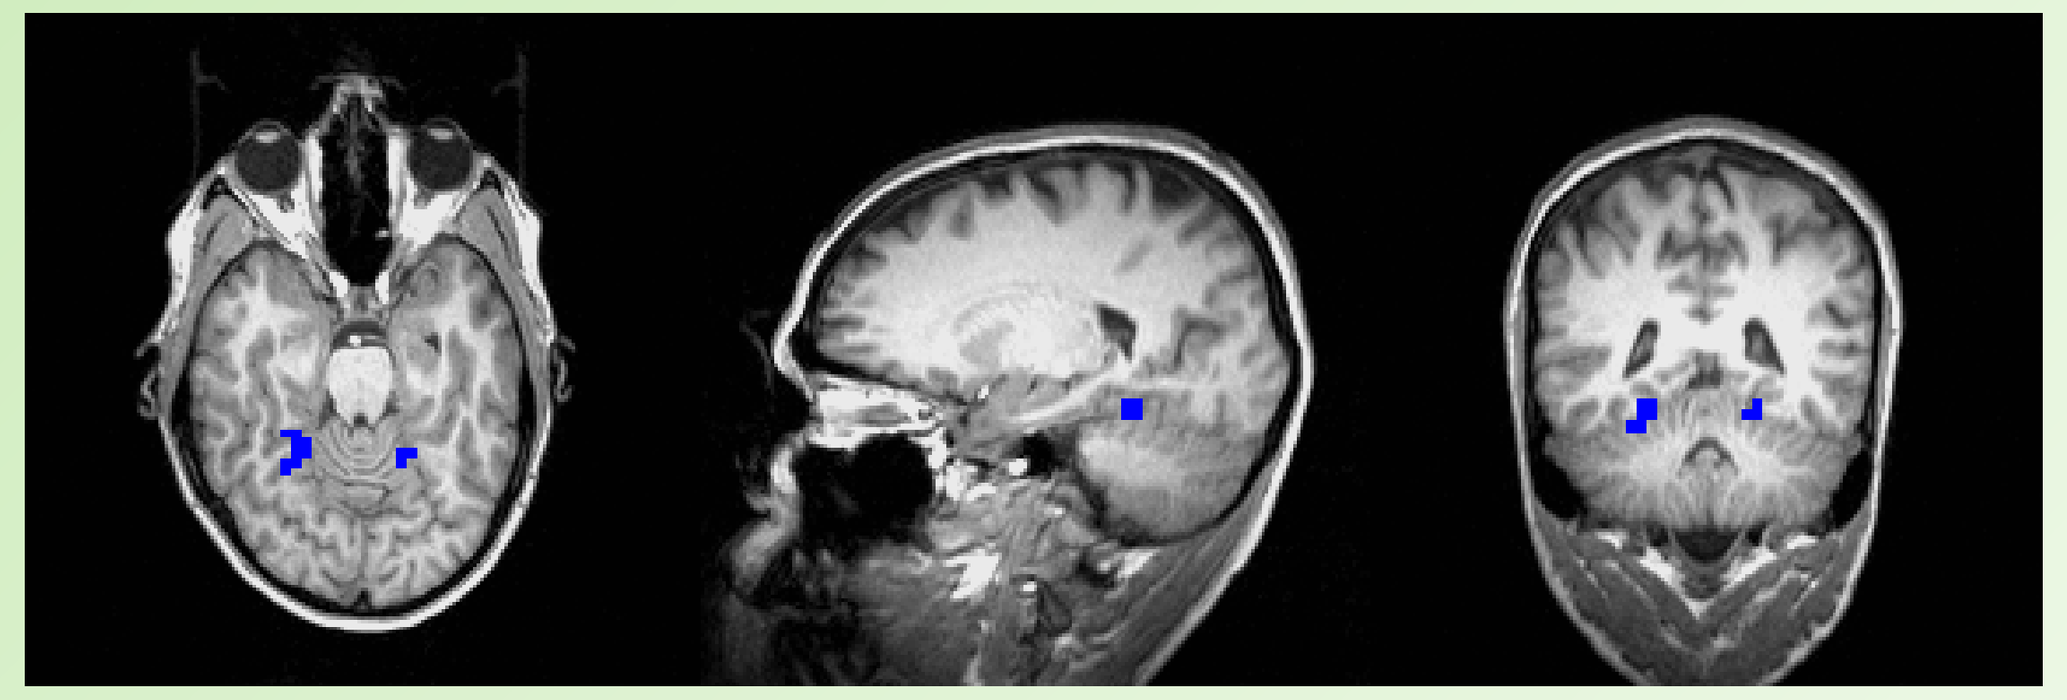

Scene category can be decoded from PPA (Walther et al., 2009).

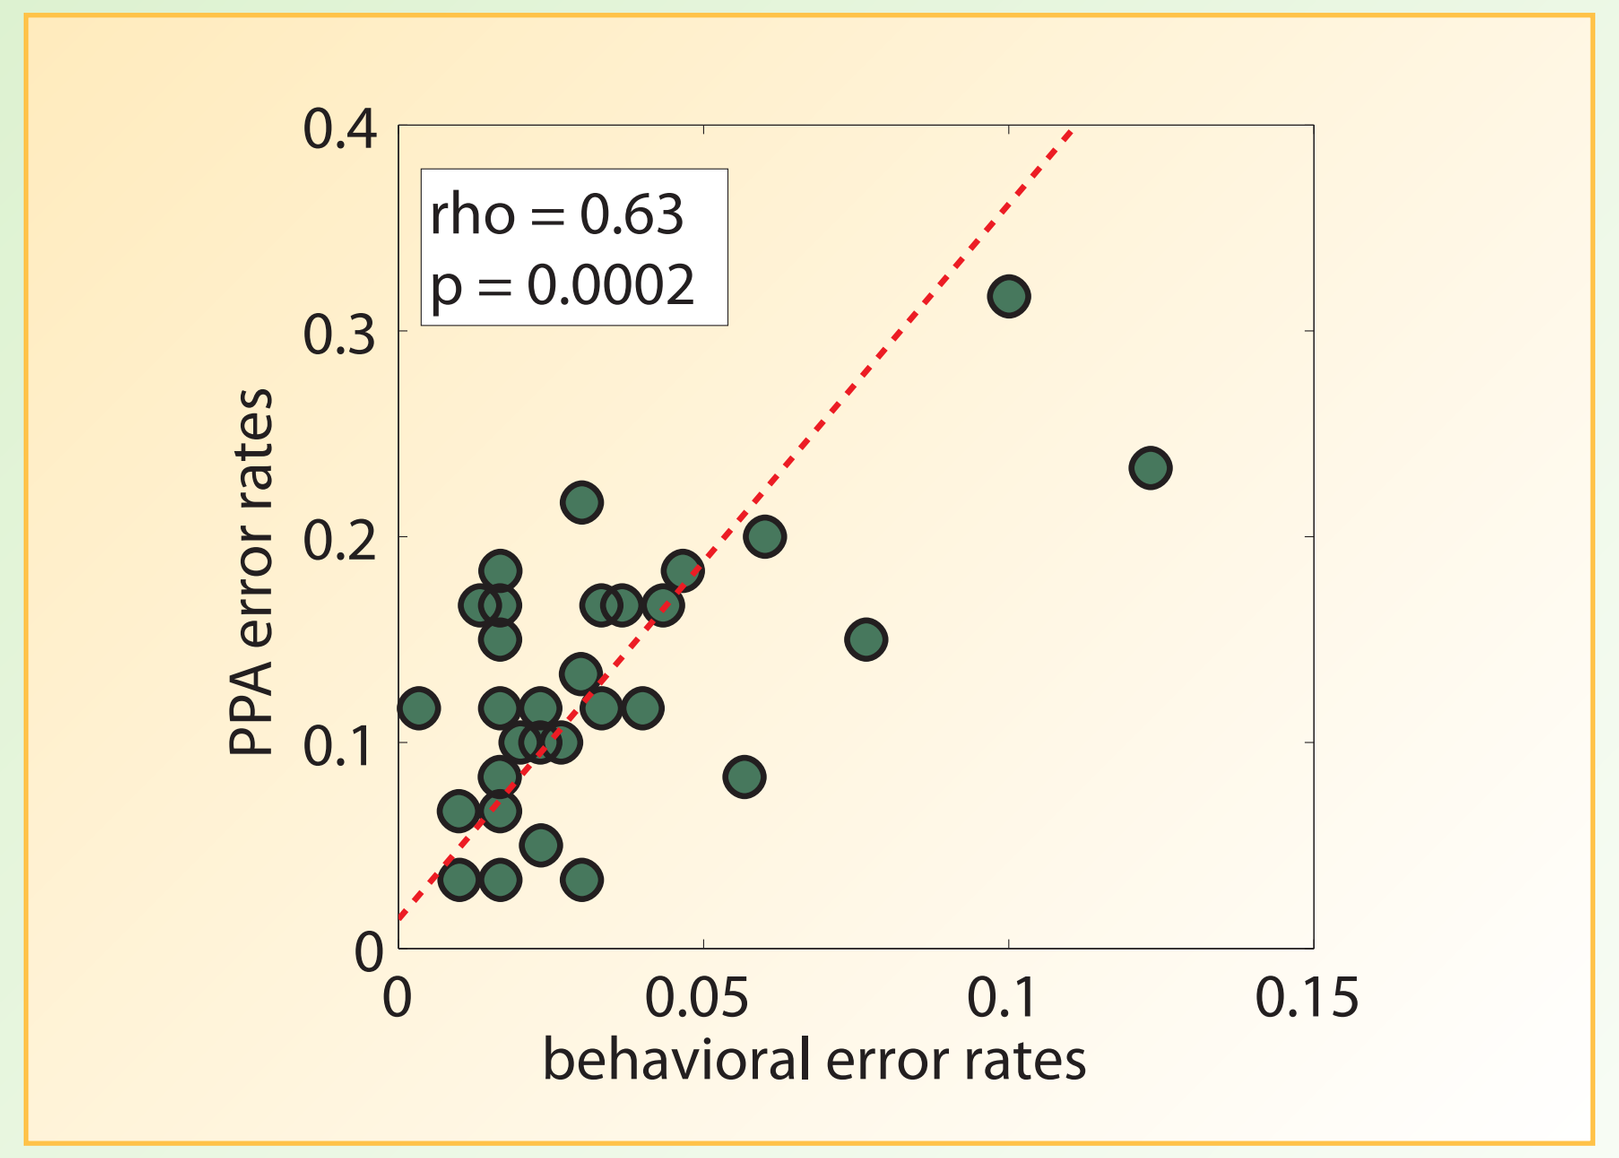
rho = 0.63
p = 0.0002
PPA error rates
behavioral error rates
0.4
0.3
0.2
0.1
0
0.05
0.1
0.15

Human scene categorization correlates best with decoding in PPA (Walther et al., 2009).

fMRI Pattern Recognition

A linear Support Vector Machine (SVM) classified the scene category presented to observers.

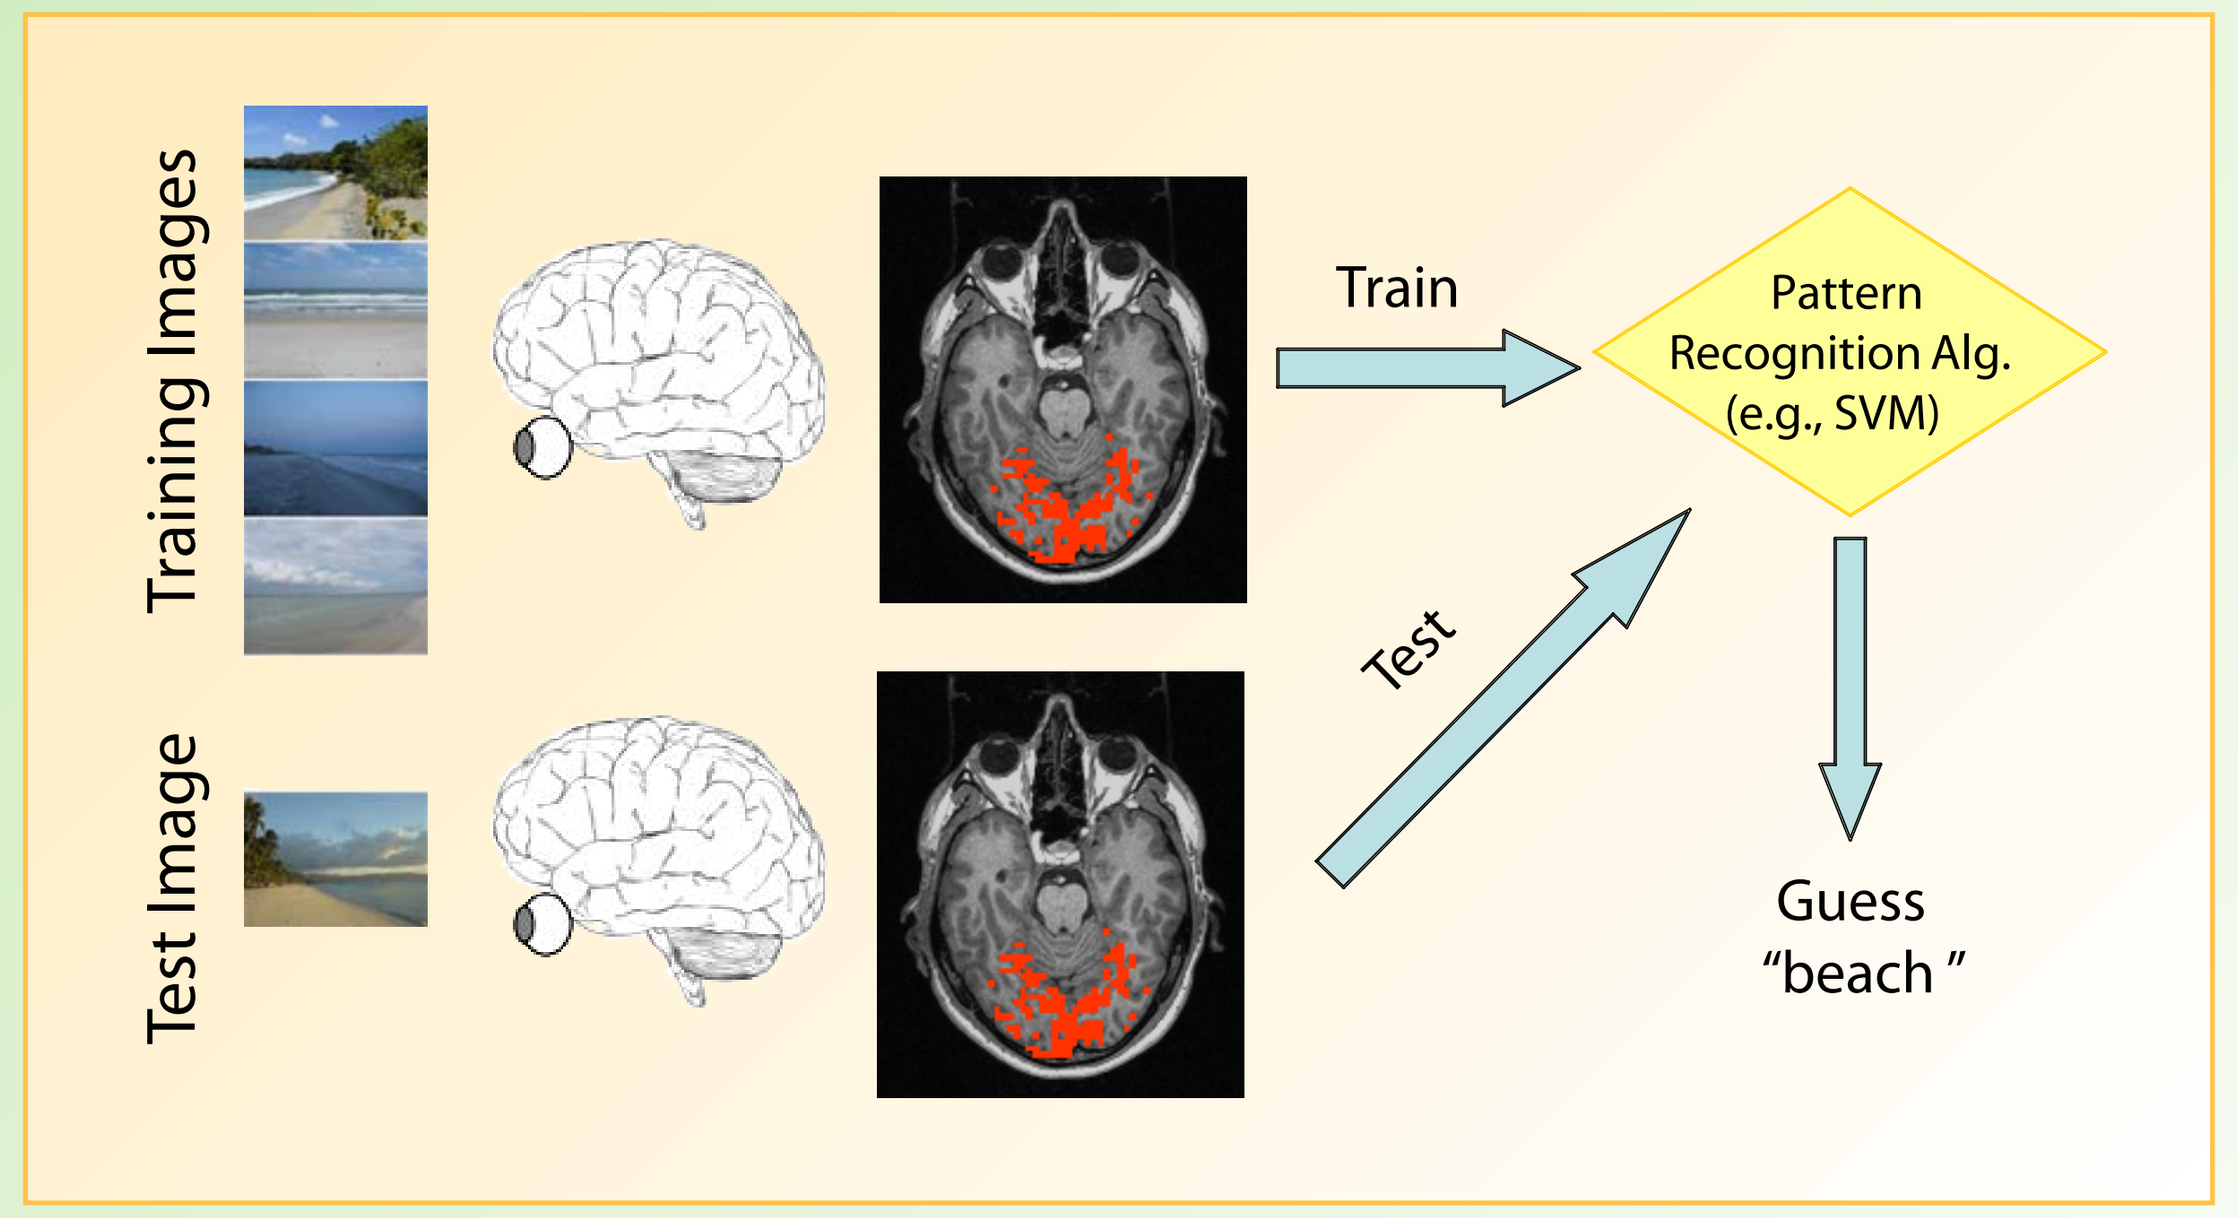
Training Images
Test Image
Train
Test
Pattern Recognition Alg. (e.g., SVM)
Guess
“beach”

12 runs were comprised of 3 groups of 4 blocks. Groups contained 1 block per scene category separated by a fixation period.

Leave-one-group-out (LOGO) cross-validation was used to assess classifier accuracy.

LOGO was performed seperately for each condition.

References

Evans, K. K., & Treisman, A. (2005). Perception of objects in natural scenes: is it really attention free? *Journal of Experimental Psychology: Human Perception and Performance, 31*(6), 1476-1492.

Li, F. F., VanRullen, R., Koch, C., & Perona, P. (2002). Rapid natural scene categorization in the near absence of attention. *Proceedings of the National Academy of Sciences of the United States of America, 99*(14), 9596-9601.

Peelen, M. V., Fei-Fei, L., & Kastner, S. (2009). Neural mechanisms of rapid natural scene categorization in human visual cortex. *Nature*, 460(7251), 94-7.

Schwartz, S., Vuilleumier, P, Hutton, C., Maravita, A., Dolan, R. J., Driver, J. (2005). Attentional Load and Sensory Competition in Human Vision: Modulation of fMRI Responses by Load at Fixation during Task-irrelevant Stimulation in the Peripheral Visual Field. *Cerebral Cortex*. 15(6), 770-786.

Walther, D. B., Caddigan, E., Fei-Fei, L., & Beck, D. M. (2009). Natural scene categories revealed in distributed patterns of activity in the human brain. *Journal of Neuroscience, 29*(34), 10573-10581.

Yi, D. J., Woodman, G. F., Widders, D., Marois, R., Chun, M. M. (2004). Neural fate of ignored stimuli: dissociable effects of perceptual and working memory load. *Nature Neuroscience*, 7(9), 992-996.

Acknowledgements

This work was funded by National Institutes of Health Grant: R01 EY019429 (CRCNS) to LFF and DB.

Task Design

Scenes were drawn from 4 categories:
Beaches, City Streets, Mountains, and Highways.

On alternating runs, participants attended scenes (subtending 24 degrees) or performed a demanding conjunction task (Schwartz et al., 2005) superimposed on the scenes at fixation.

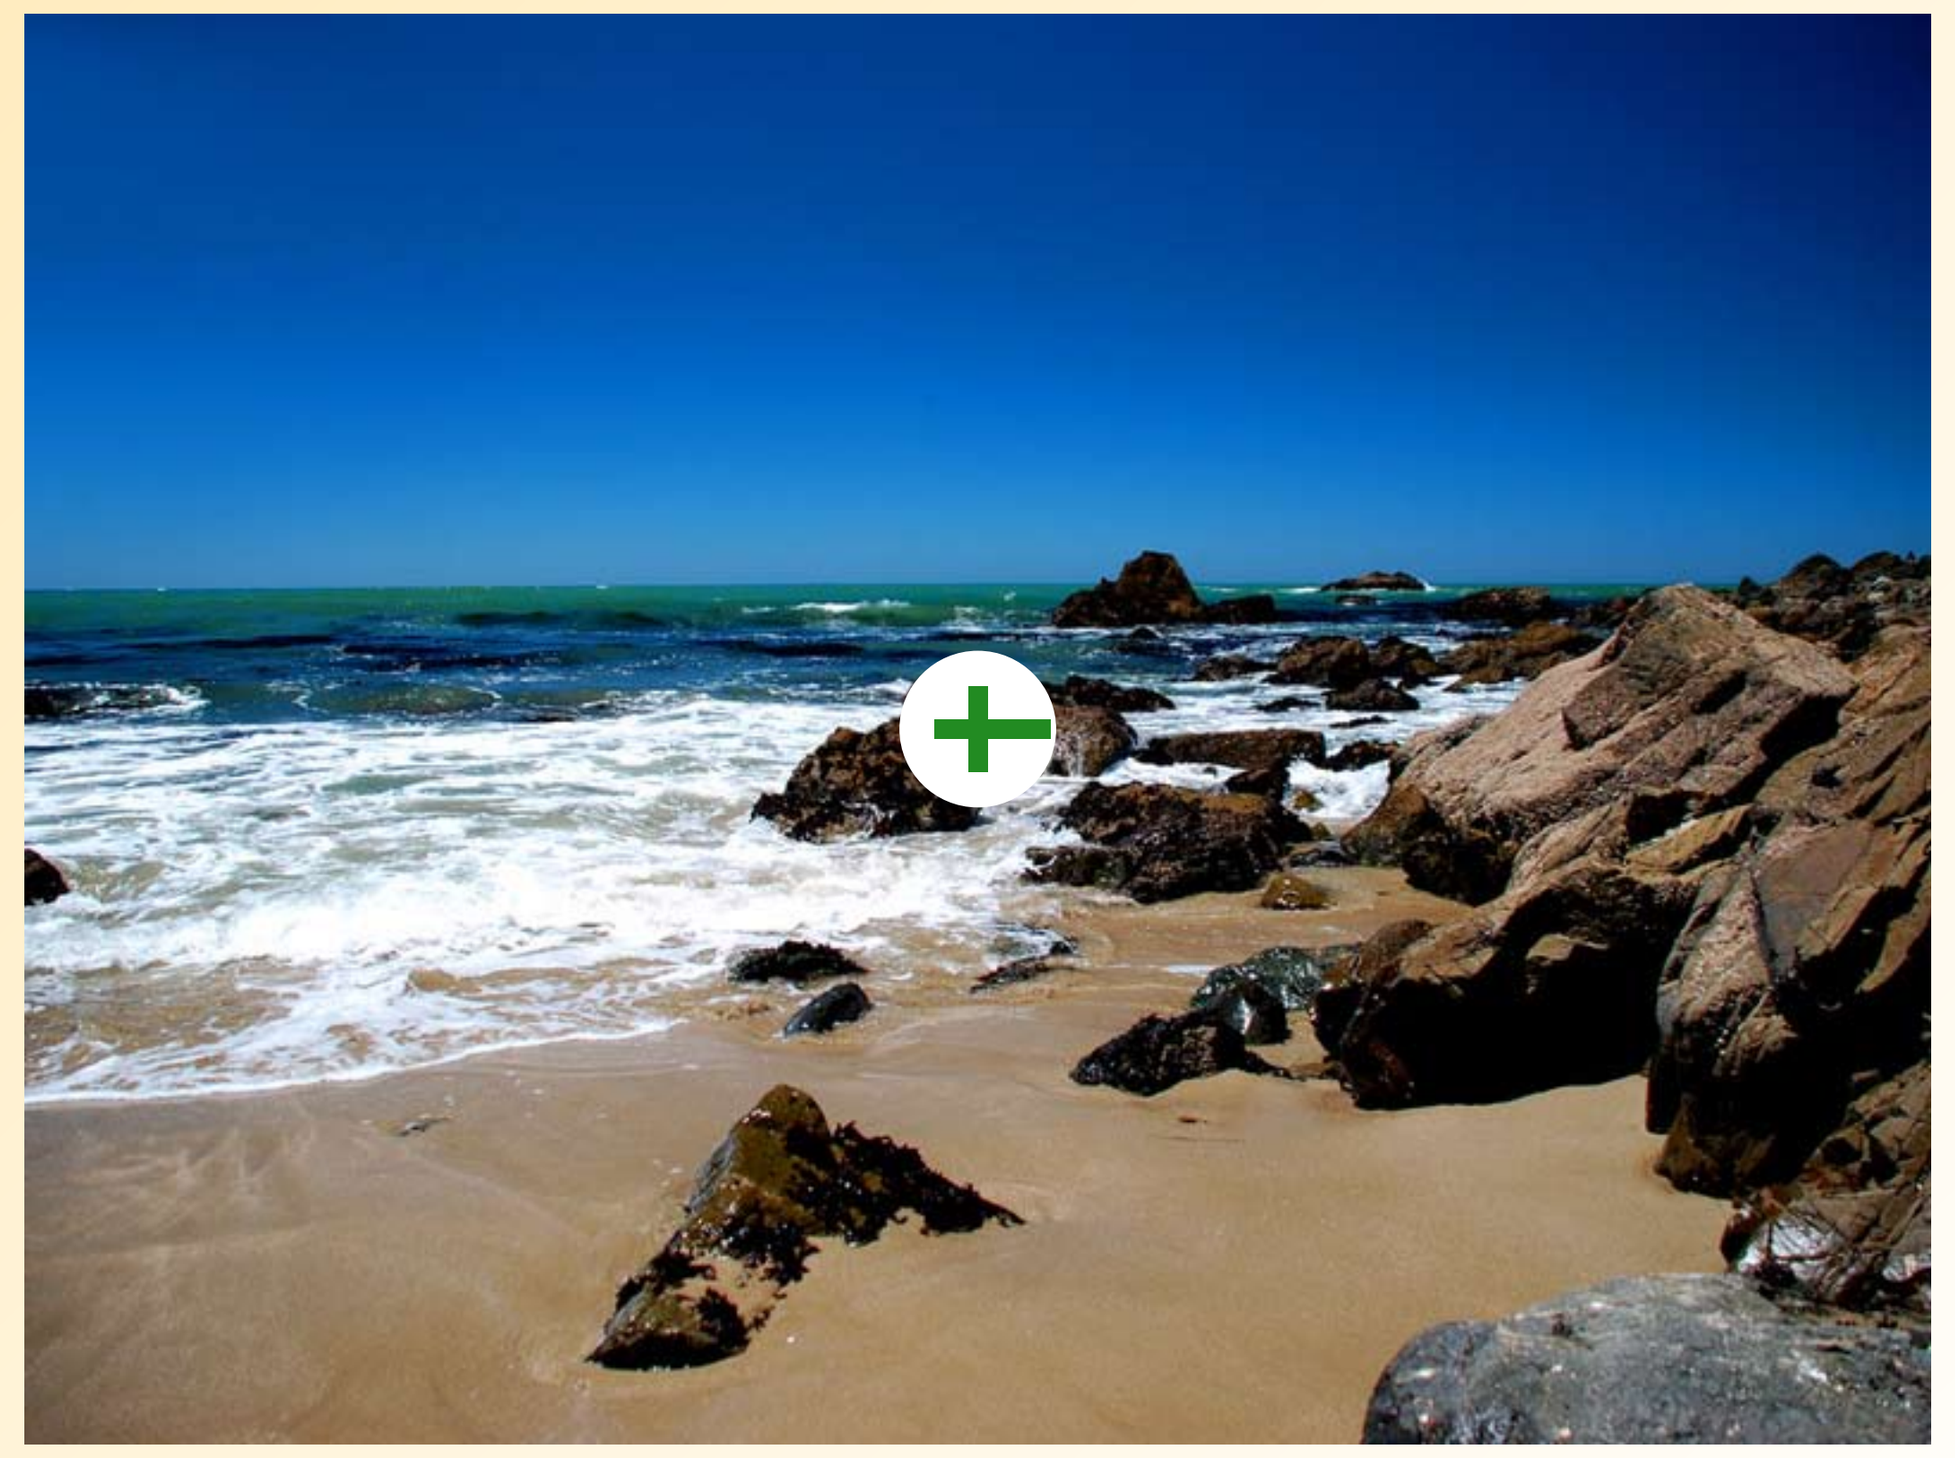

Scene category exemplars were presented with a fixation cross superimposed. Stimuli were identical in both conditions.

Scene Task

Blocks consisted of 7 images (one image per TR; TR = 1750 ms).

Participants responded to repeats of the first image in the block (no other images repeated).

Targets occurred 0, 1, or 2 times.

Fixation Task

16 colored crosses appeared sequentially at fixation, each pointing left or right (subtending 2 degrees).

Participants performed a conjunction search, responding to yellow left-pointing crosses and green right-pointing crosses.

The number of targets in each block was identical to the scene task.

Results

Participants performed equally well at the scene and fixation tasks.

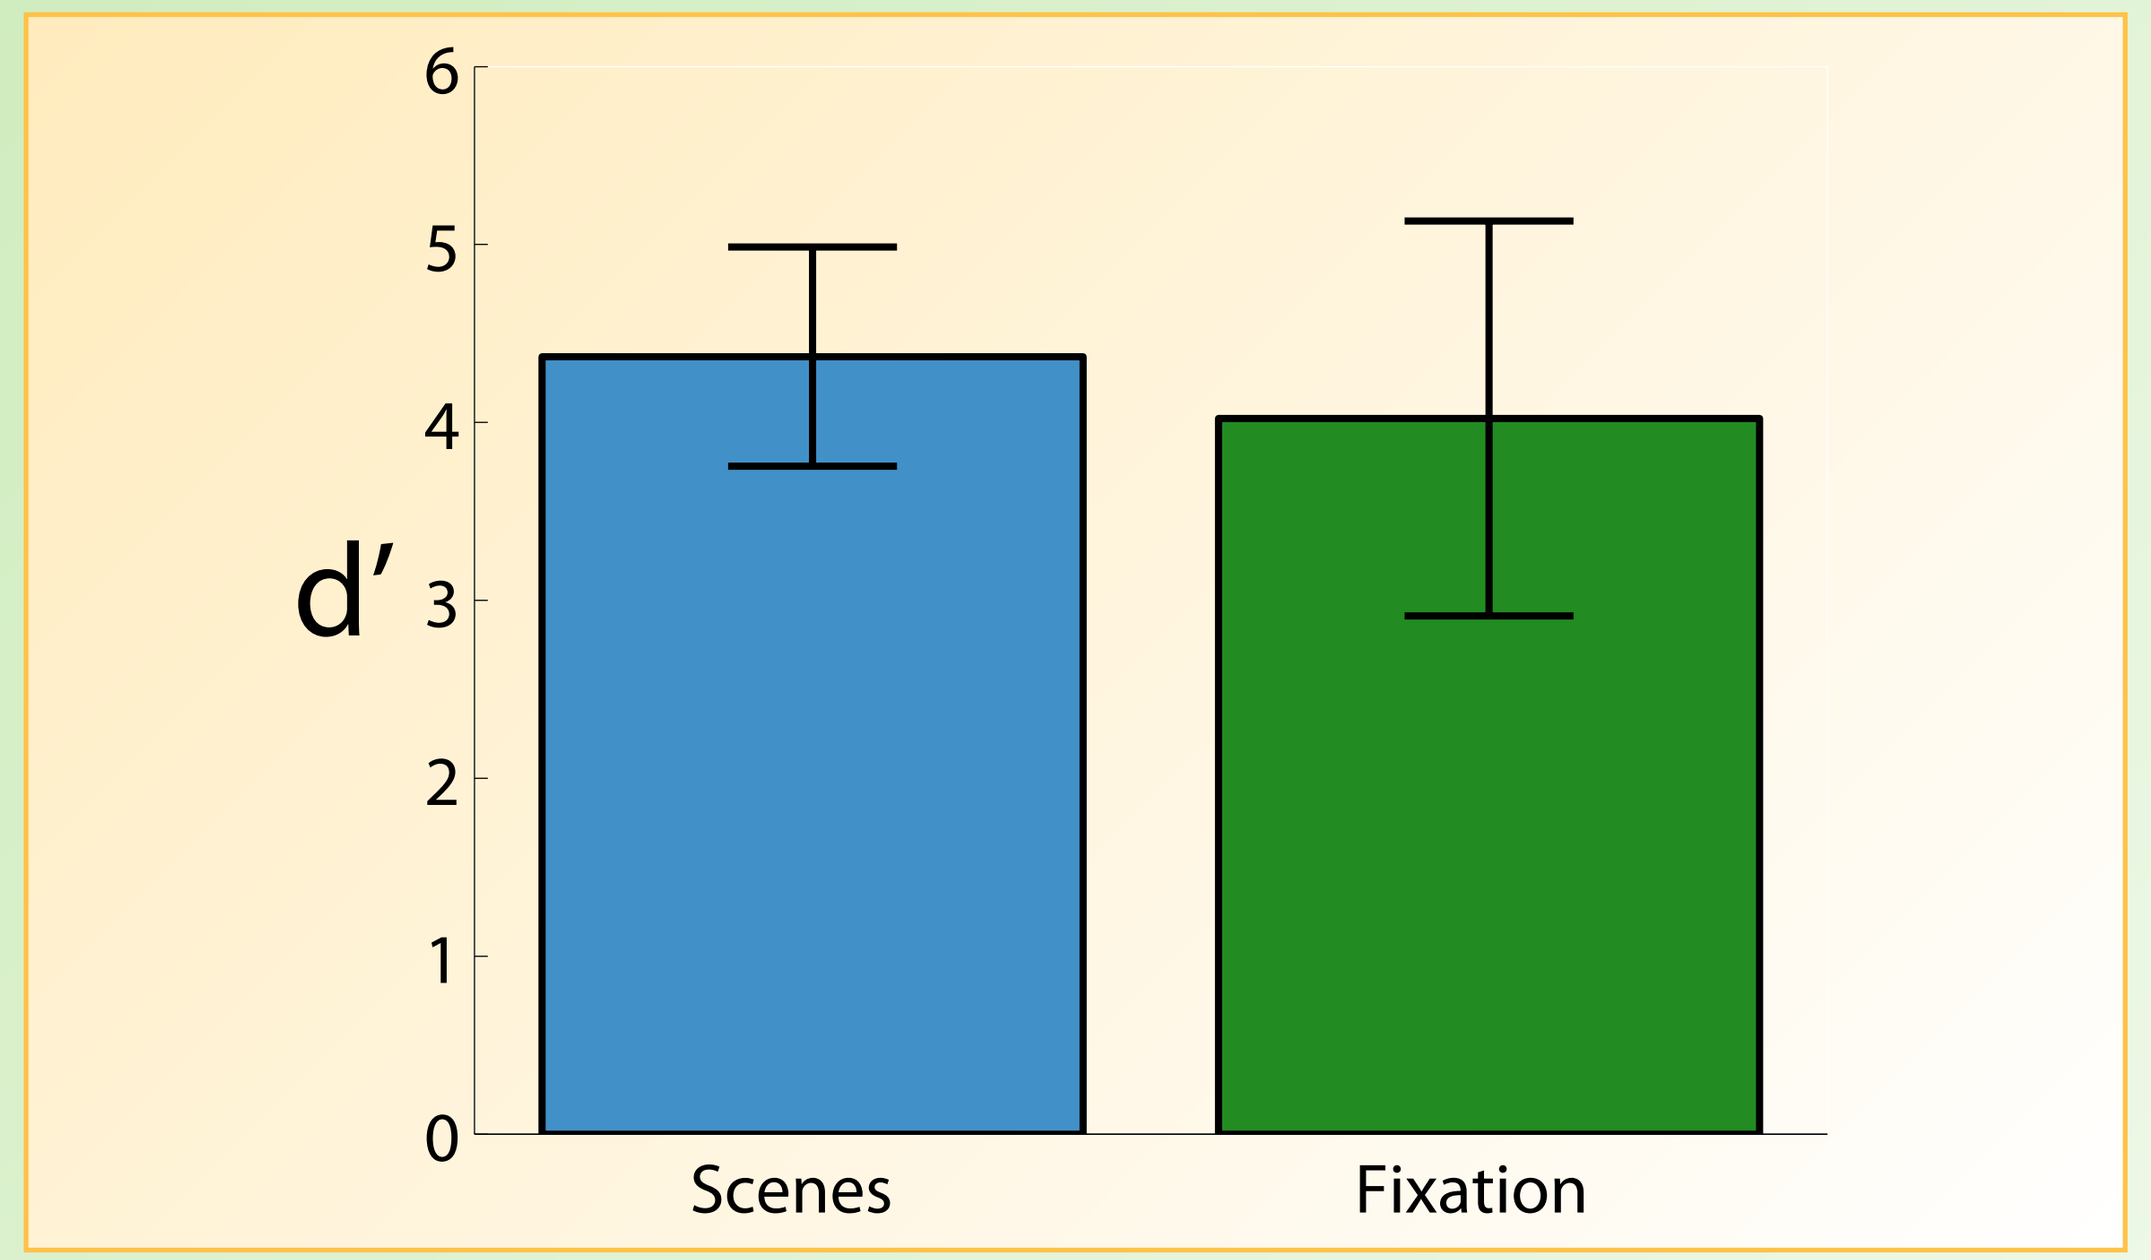
d′
6
5
4
3
2
1
0
Scenes
Fixation

Scene category decoding was significantly above chance, even during the fixation task.

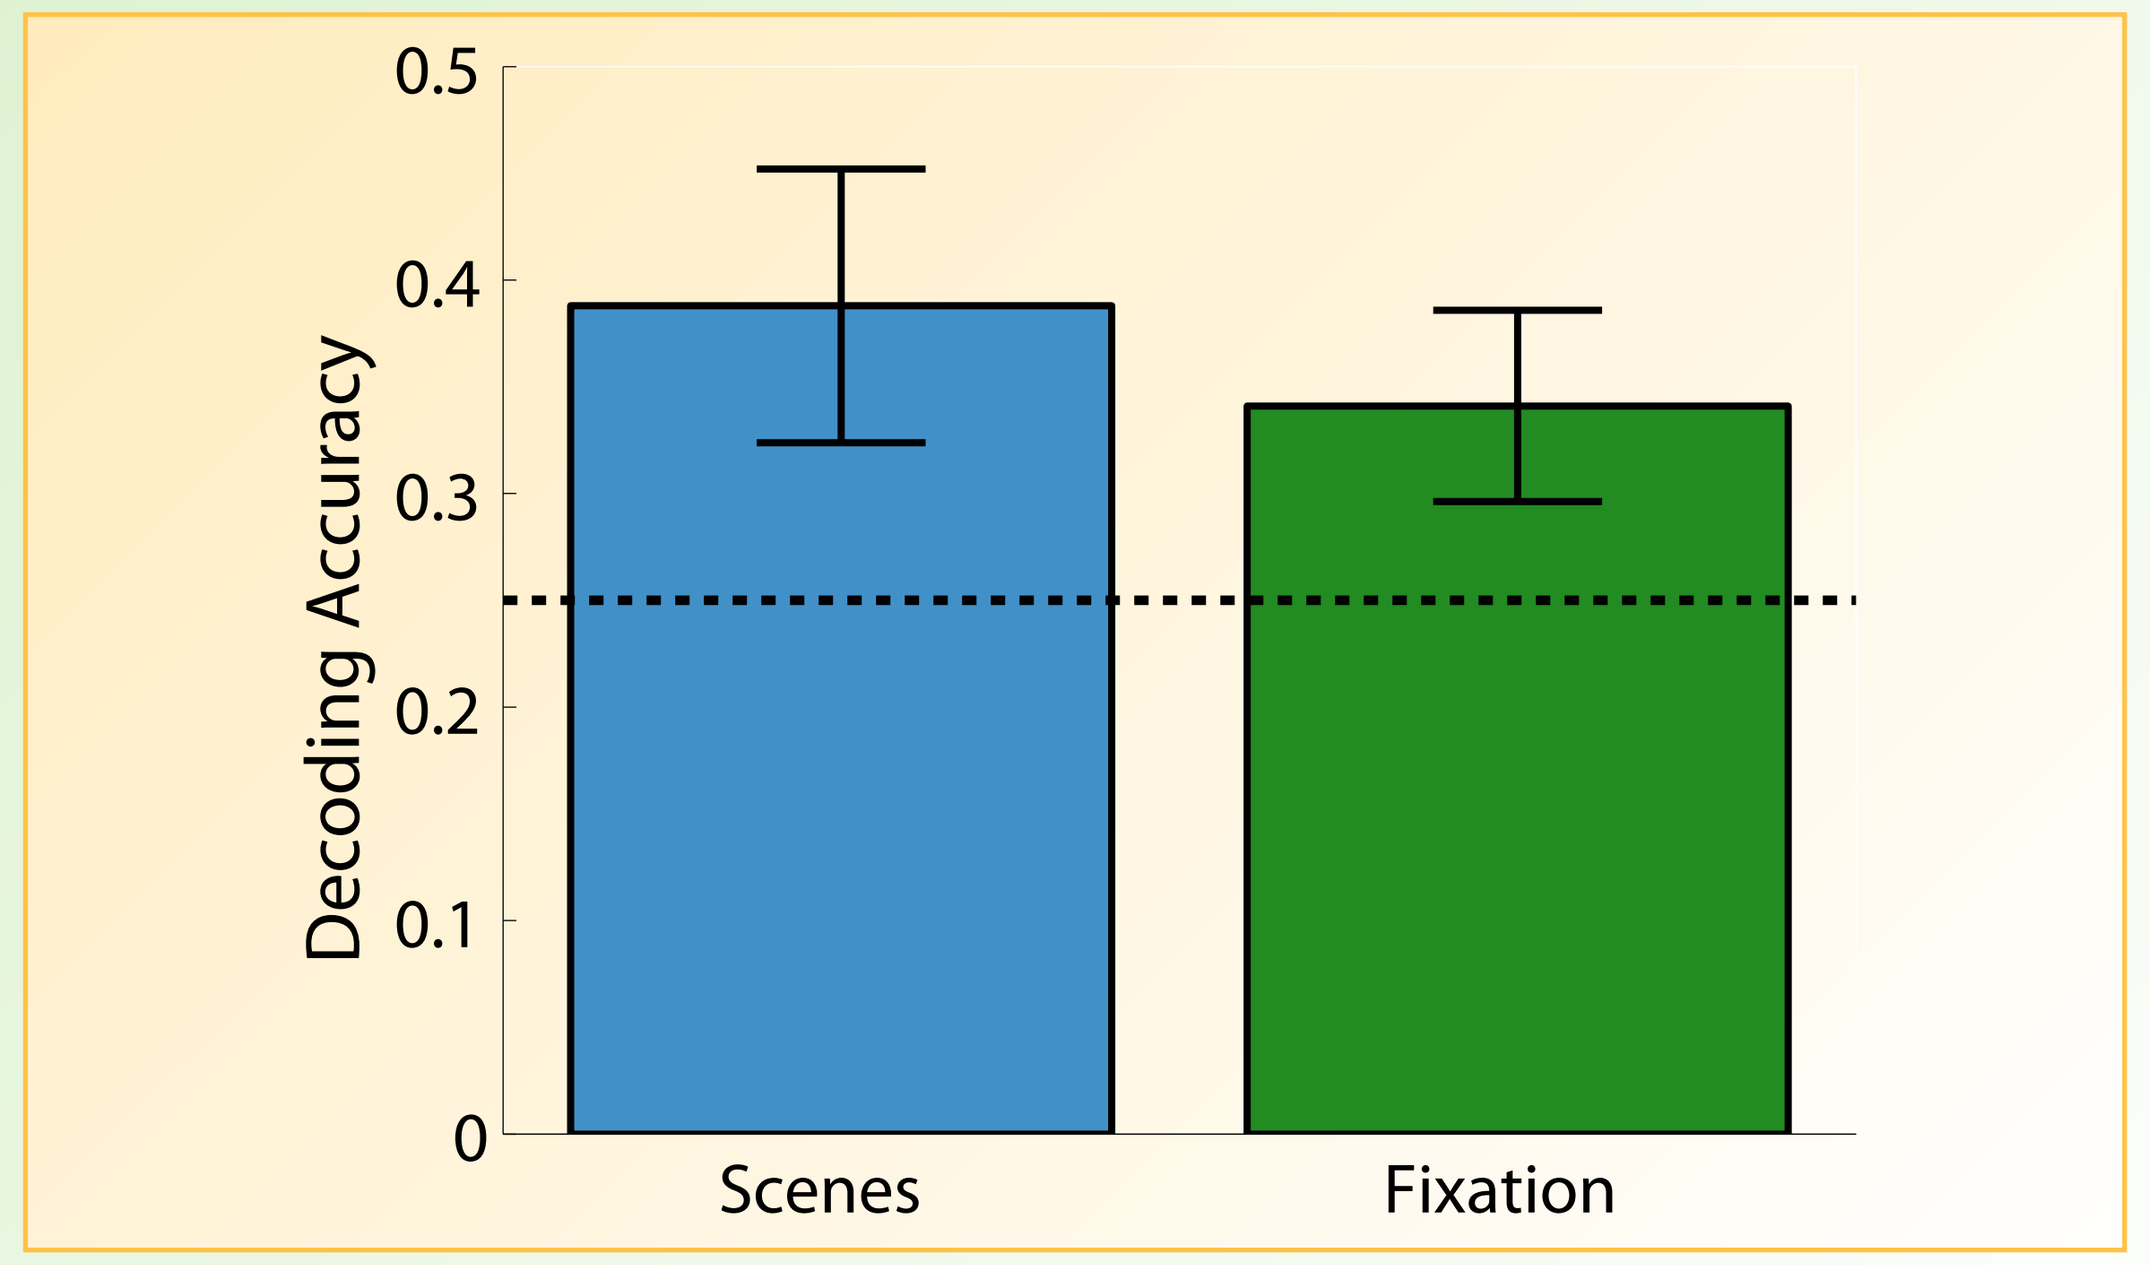
Decoding Accuracy
0.5
0.4
0.3
0.2
0.1
0
Scenes
Fixation

Decoding accuracy was significantly higher when participants attended scenes vs. the fixation task.

Conclusions

Scene category information is represented when observers’ attention is directed elsewhere.

Attention improves the representation of natural scene category information

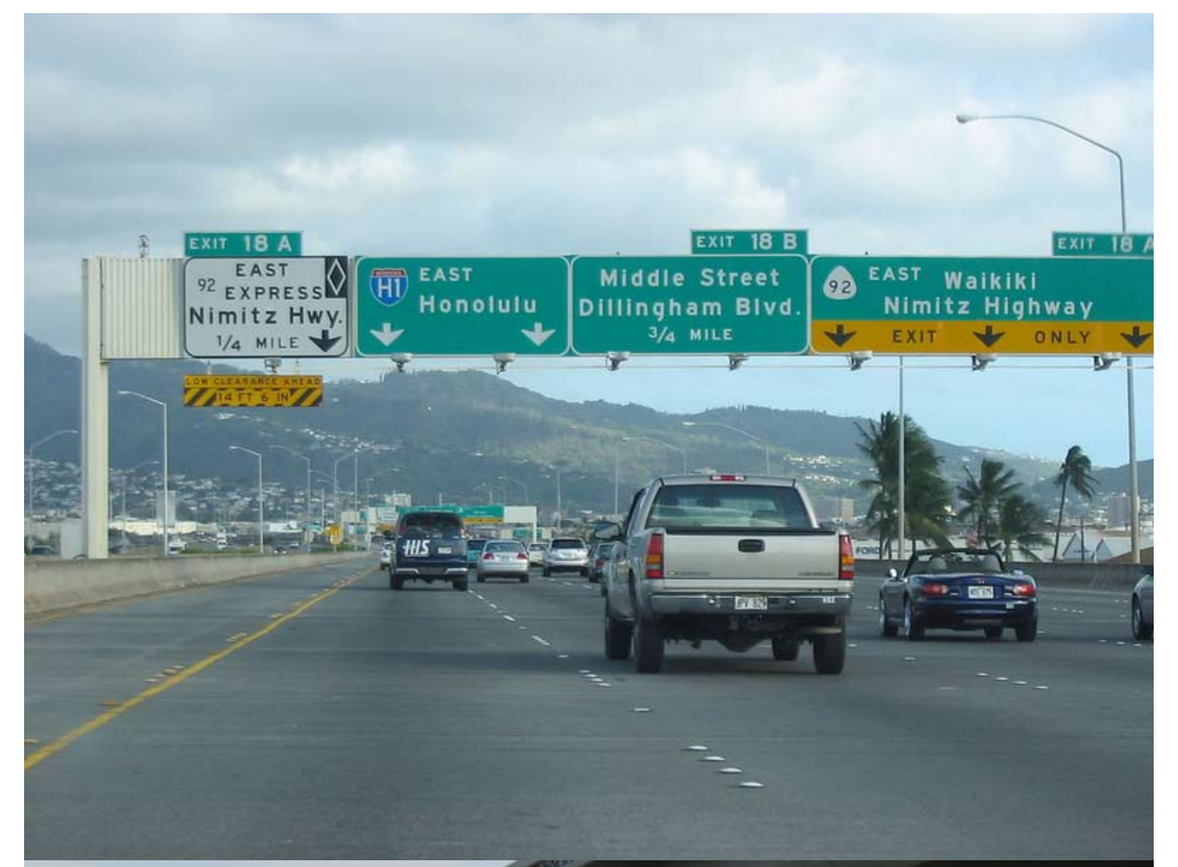
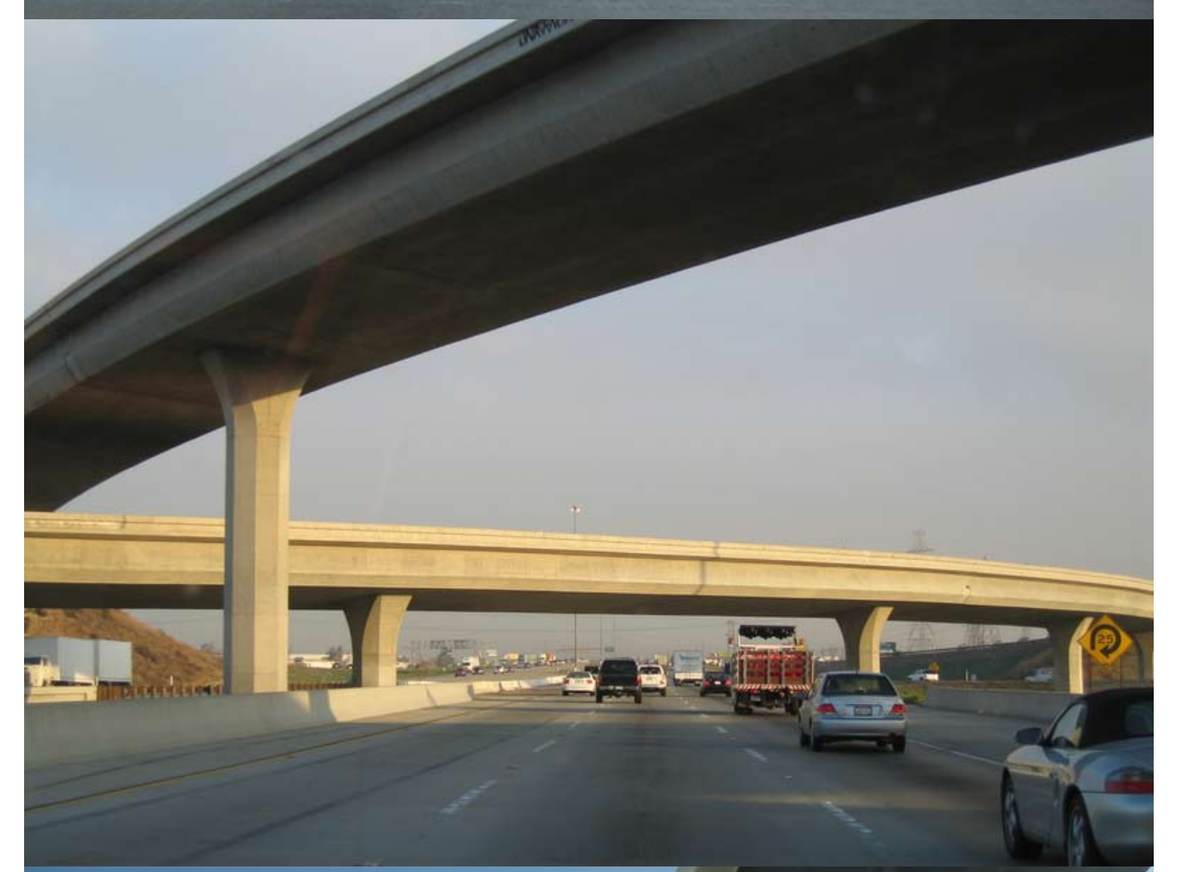
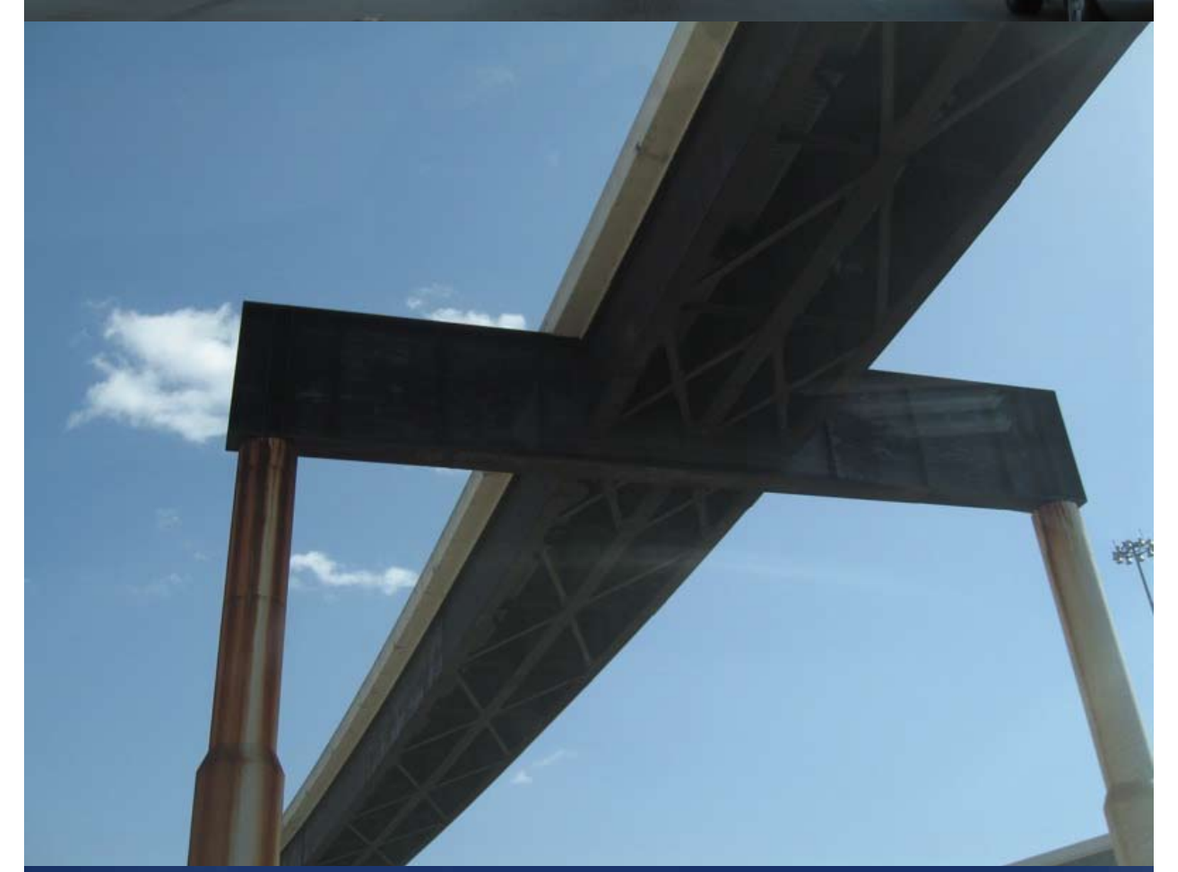
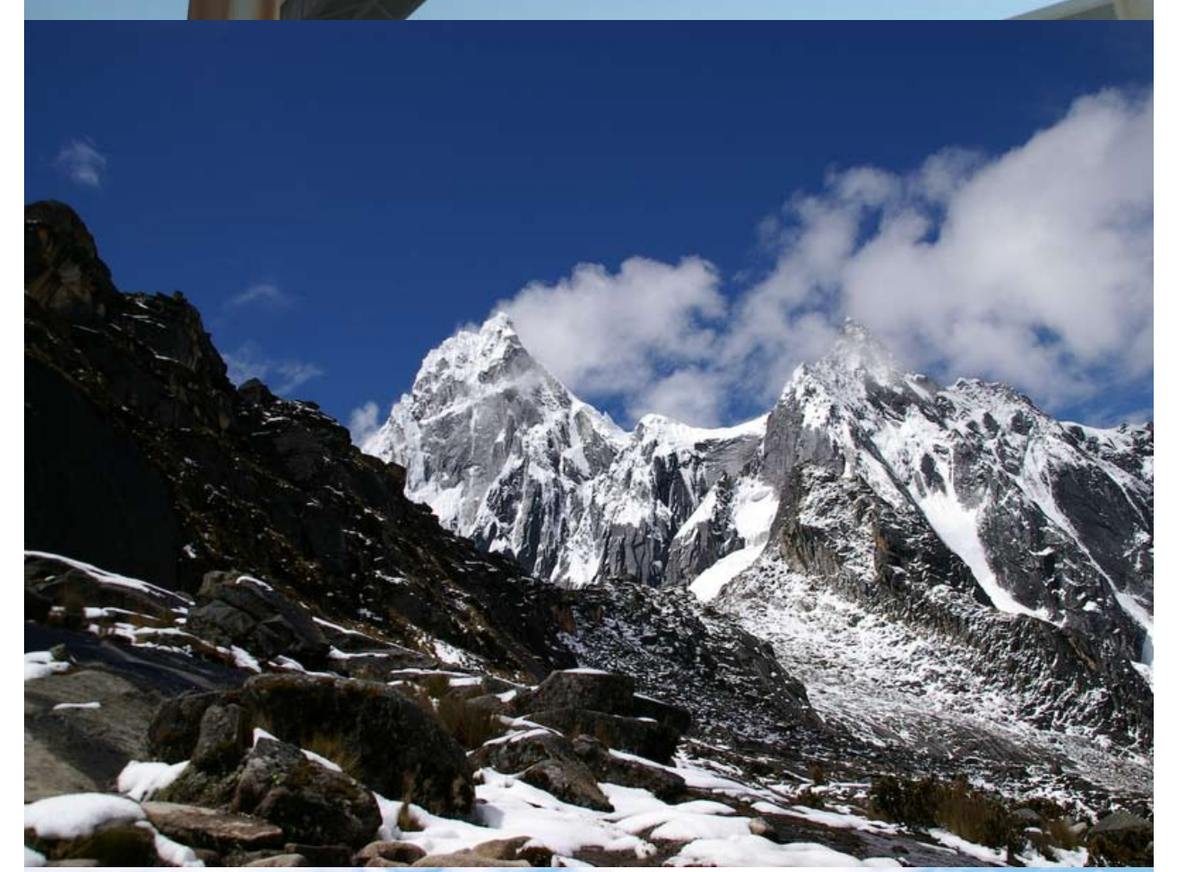
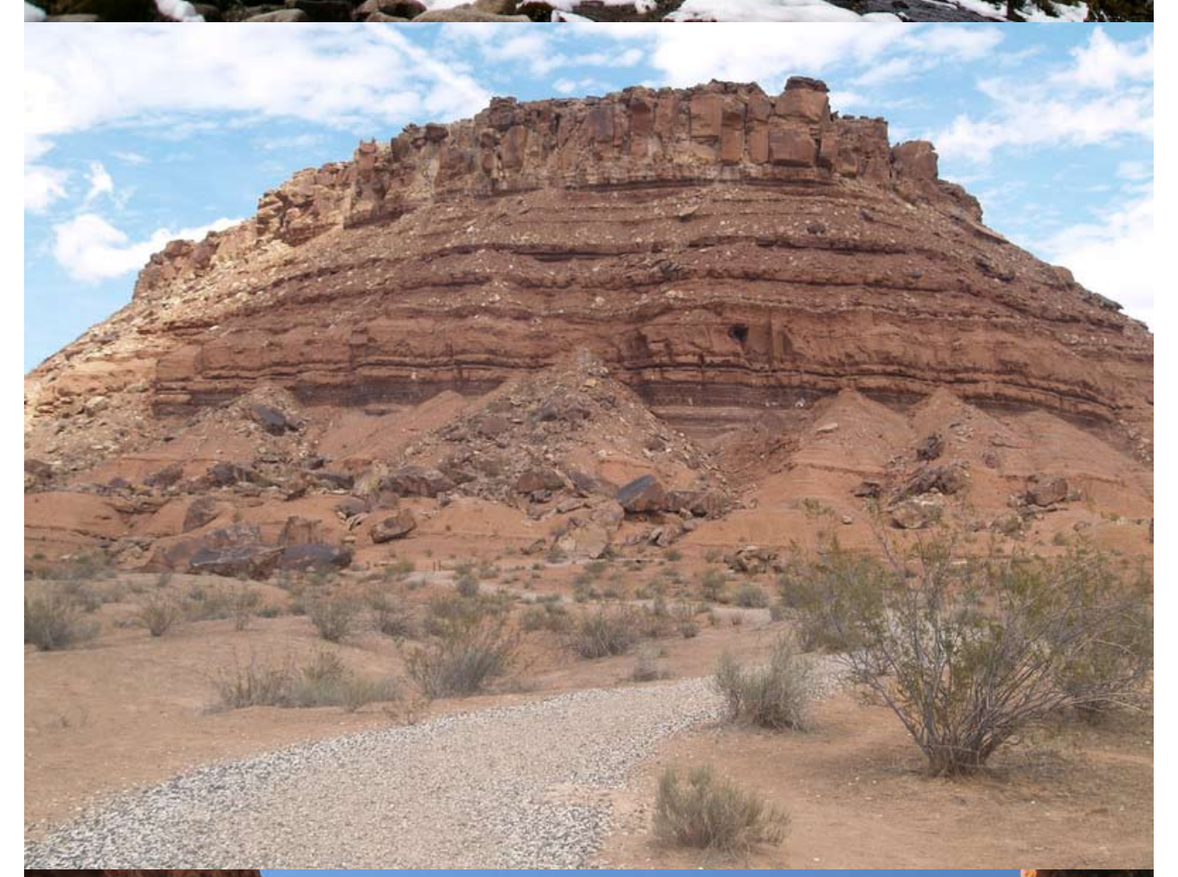
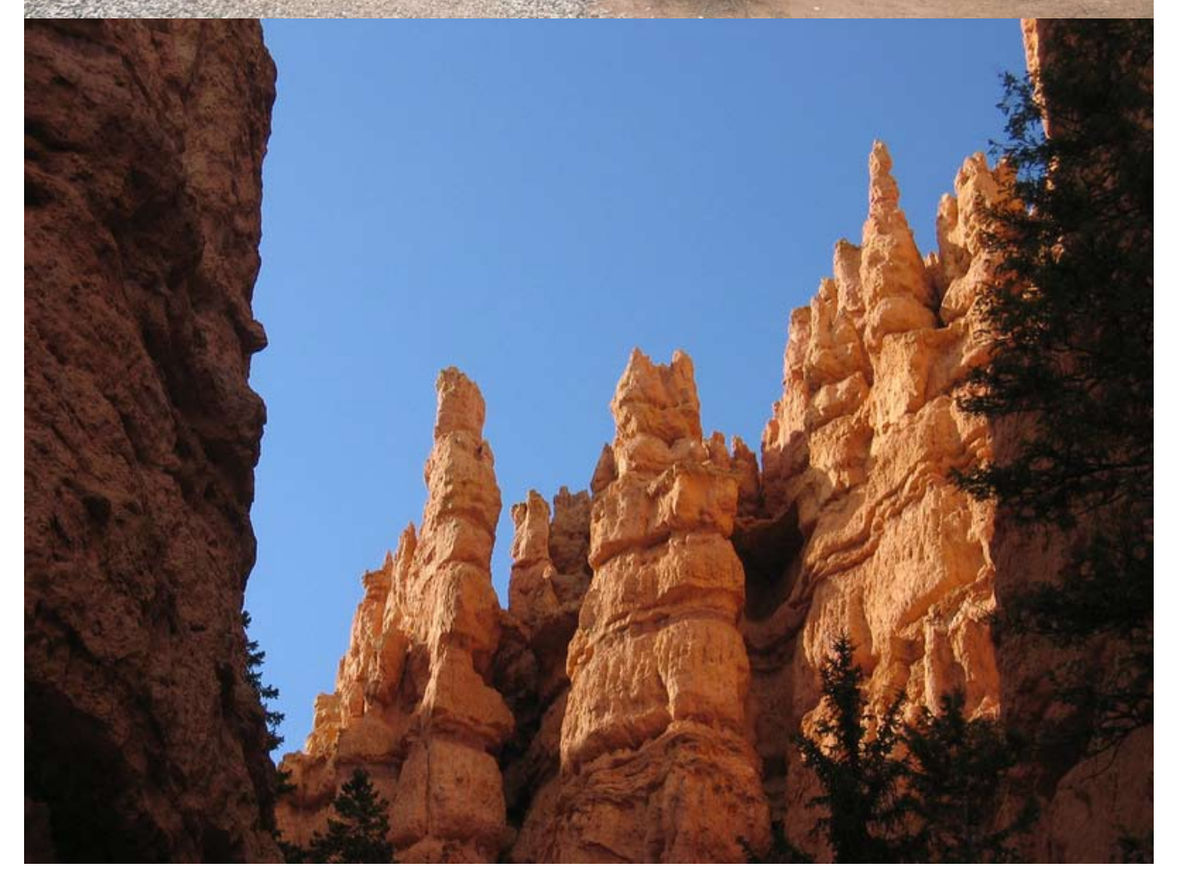